Cardiovascular effects of methamphetamine, 3,4-methylenedioxymethamphetamine, and other entactogens in C57BL/6 and BTBR T+Itpr3tf/J mice.

Holloway KH[1], Shaw HE[1]; Kaur H[2] and Fantegrossi WE[1]

1-Department of Pharmacology and Toxicology, University of Arkansas for Medical Sciences, Little Rock, AR;
2-PharmAla Biotech, Toronto, Ontario, Canada

UAM College of Medicine

INTRODUCTION

- There is renewed interest in potential therapeutic effects of psychedelic drugs for a range of neuropsychiatric conditions, including psychedelic-like entactogens such as MDMA.
- Hypertensive effects of MDMA have been demonstrated in multiple species, and these effects may be treatment-limiting in a therapeutic context.
- MDMA and its analogues are being explored as potential treatments for social withdrawal secondary to Autism spectrum disorder (ASD), but the effects of these drugs in the BTBR T+Itpr3tf/J (BTBR) mouse – the "gold standard" model for ASD research – have not previously been characterized.
- These studies used a tail-cuff volume pressure recording system to simultaneously monitor changes in systolic and diastolic pressure elicited by injection of saline, S-methamphetamine (S-METH), racemic MDMA, 5-EAPB or 6-EAPB in restrained C57 and BTBR mice.

METHODS

- Mice were habituated to restraint, saline injection (IP), and tail cuff pressure monitoring for at least three days before drug administration.

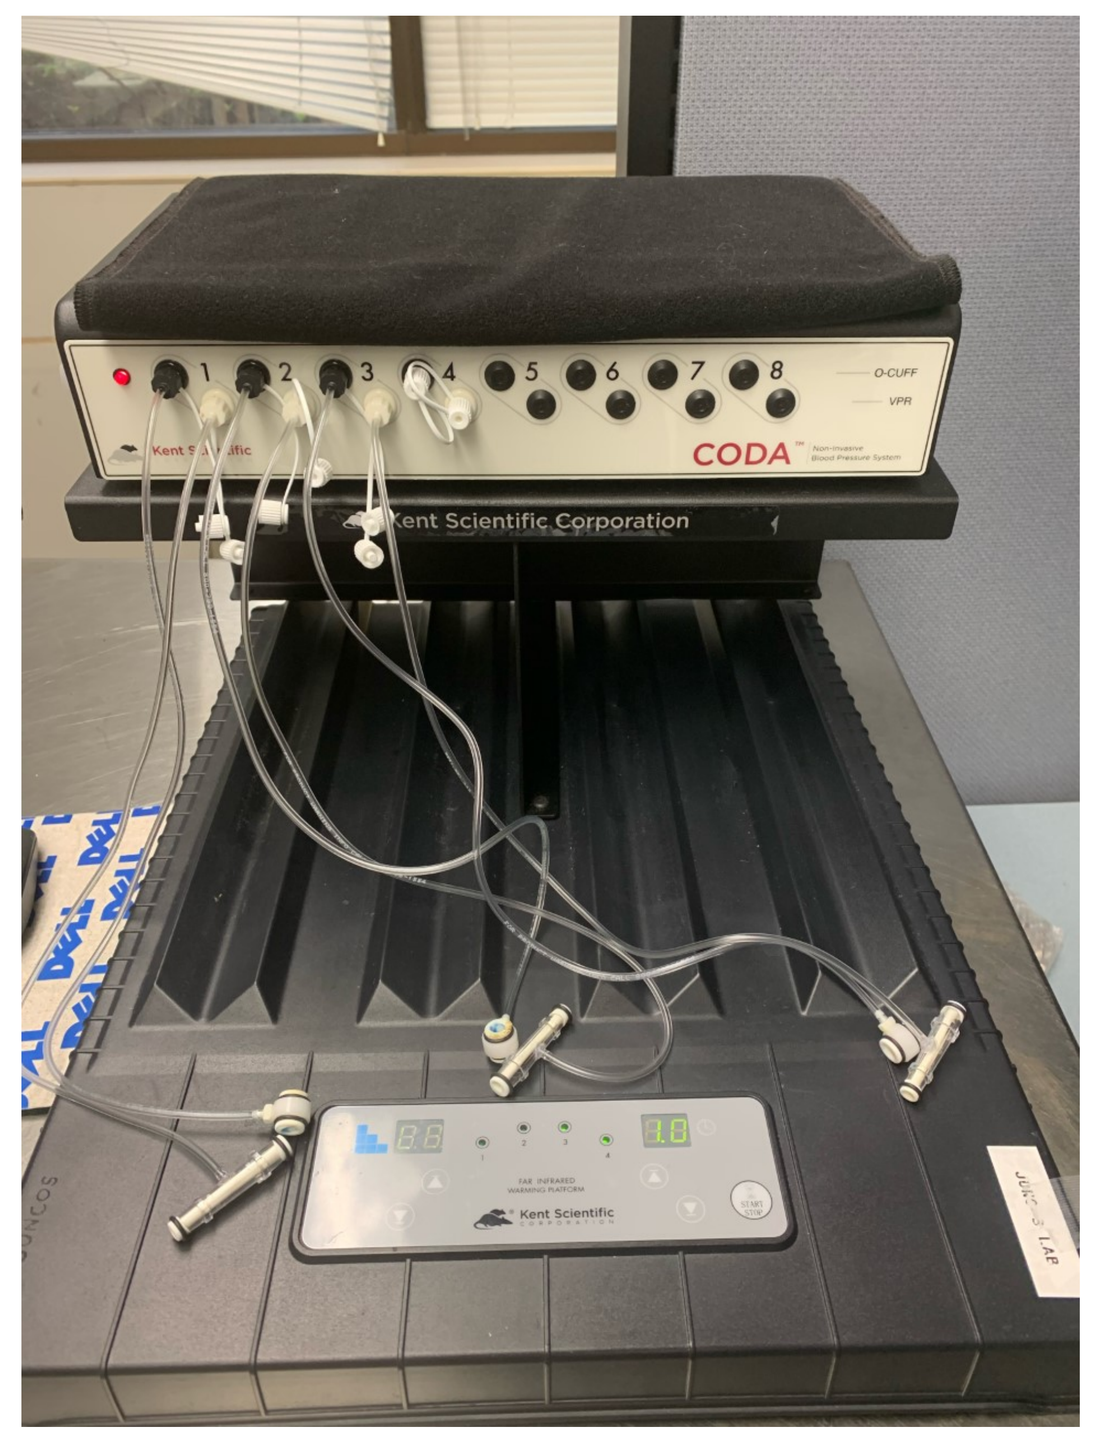

CODA
Kent Scientific Corporation

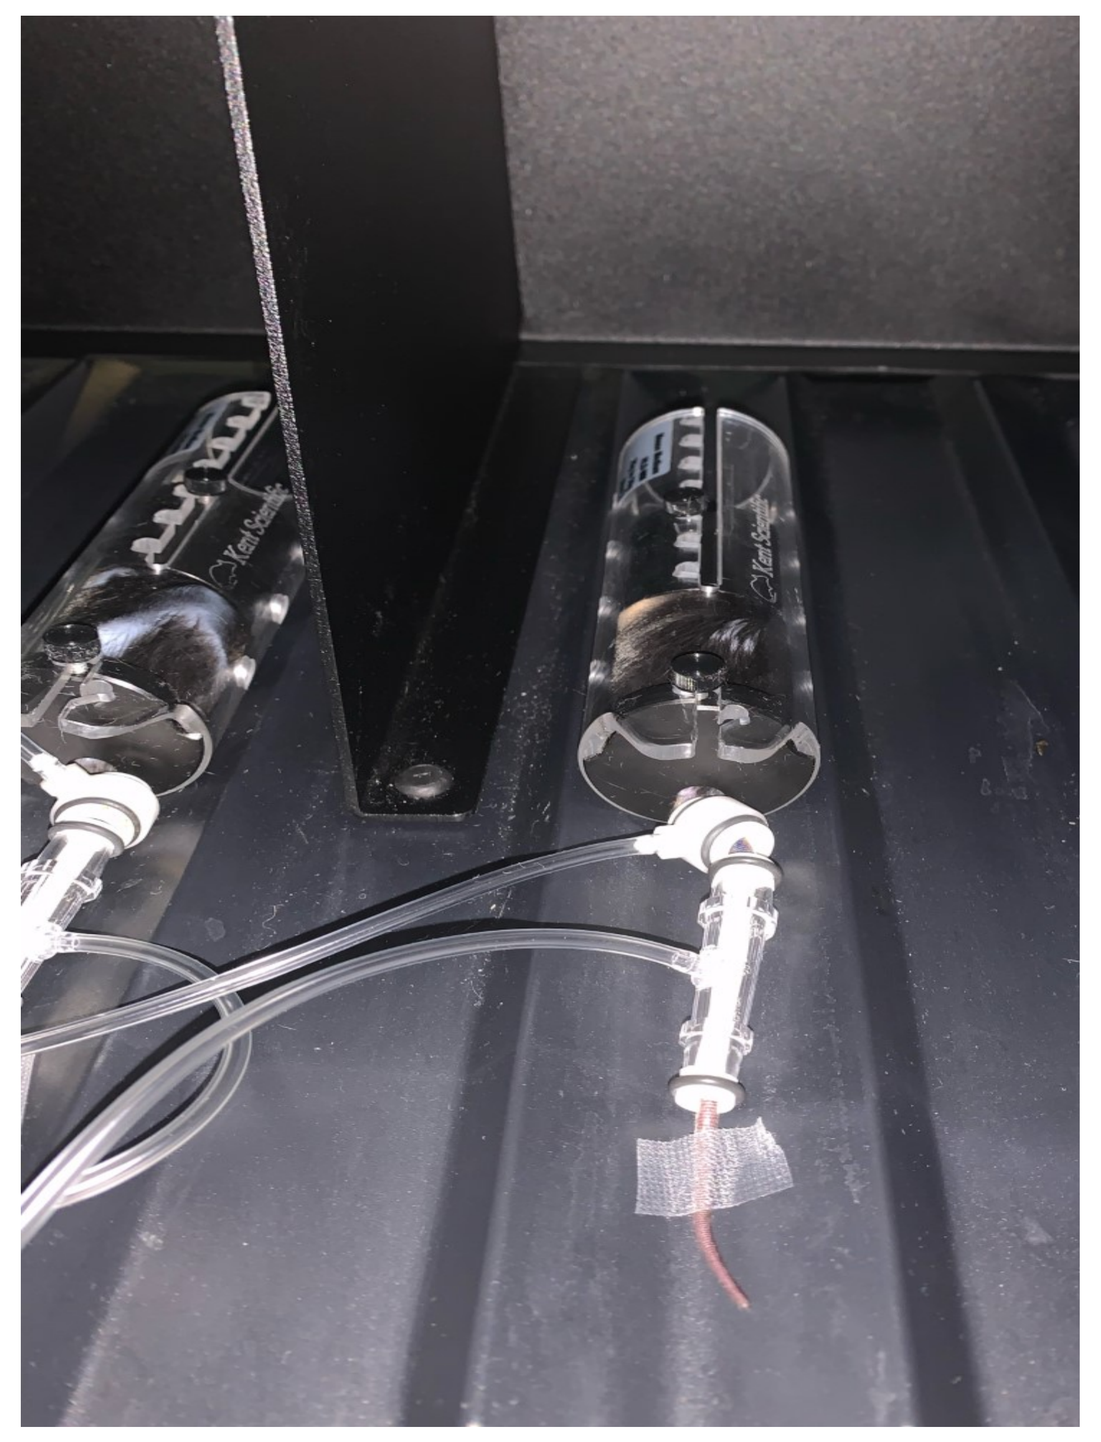

- Across days, groups of habituated mice were injected IP with increasing doses of MDMA, its enantiomers, ALA-002, METH, or the MDMA-like compounds 5-EAPB and 6-EAPB, then returned to the home cage for 30 min.
- Tail cuff pressure monitoring then determined effects on systolic, diastolic, and mean arterial pressure in restrained C57 and BTBR mice in sessions lasting ~30 min.

DRUGS

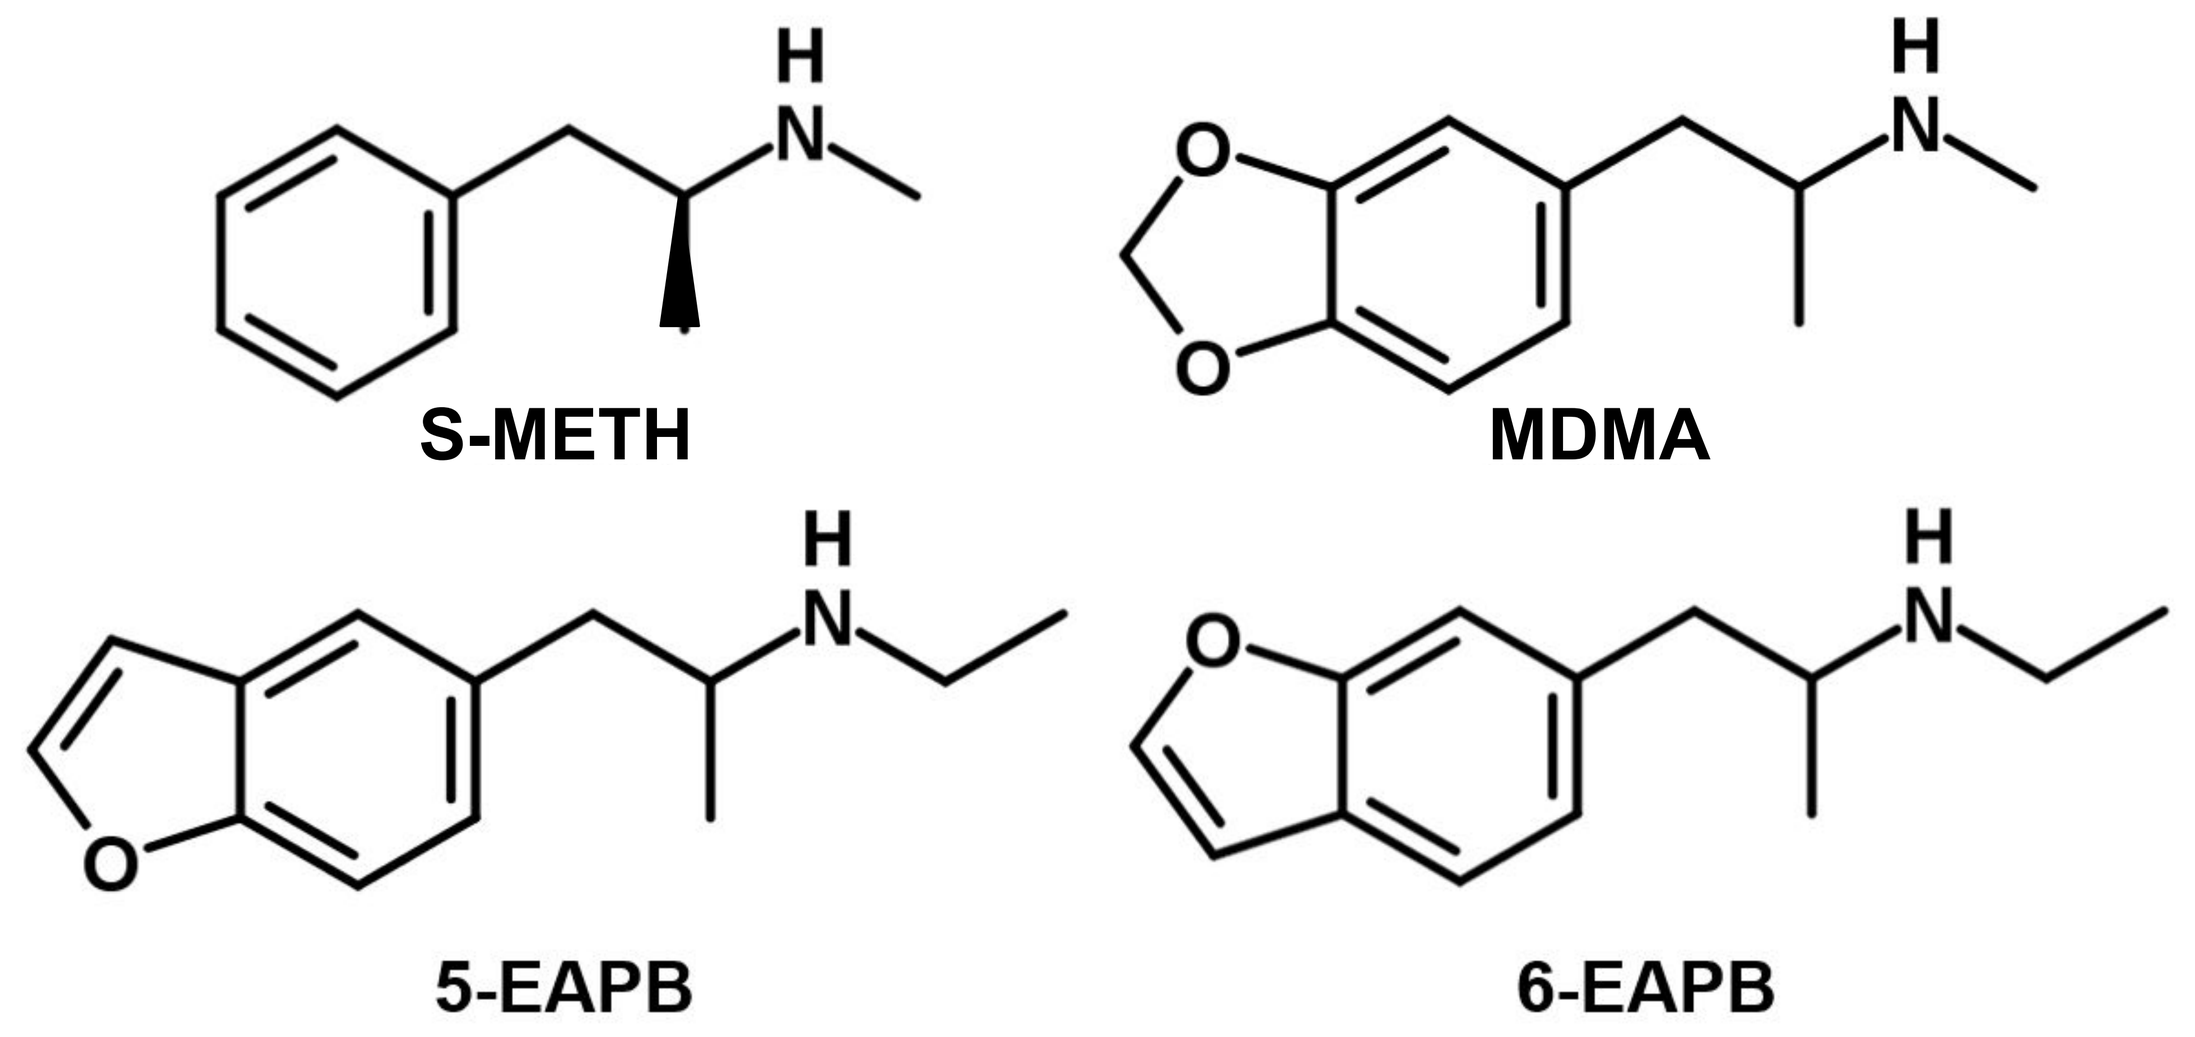

S-METH
MDMA
5-EAPB
6-EAPB

- All drugs were formulated as HCl salts and dissolved in saline prior to injection. METH was administered as the S-enantiomer, but all other drugs were injected as racemic mixtures.

EFFECTS OF RESTRAINT

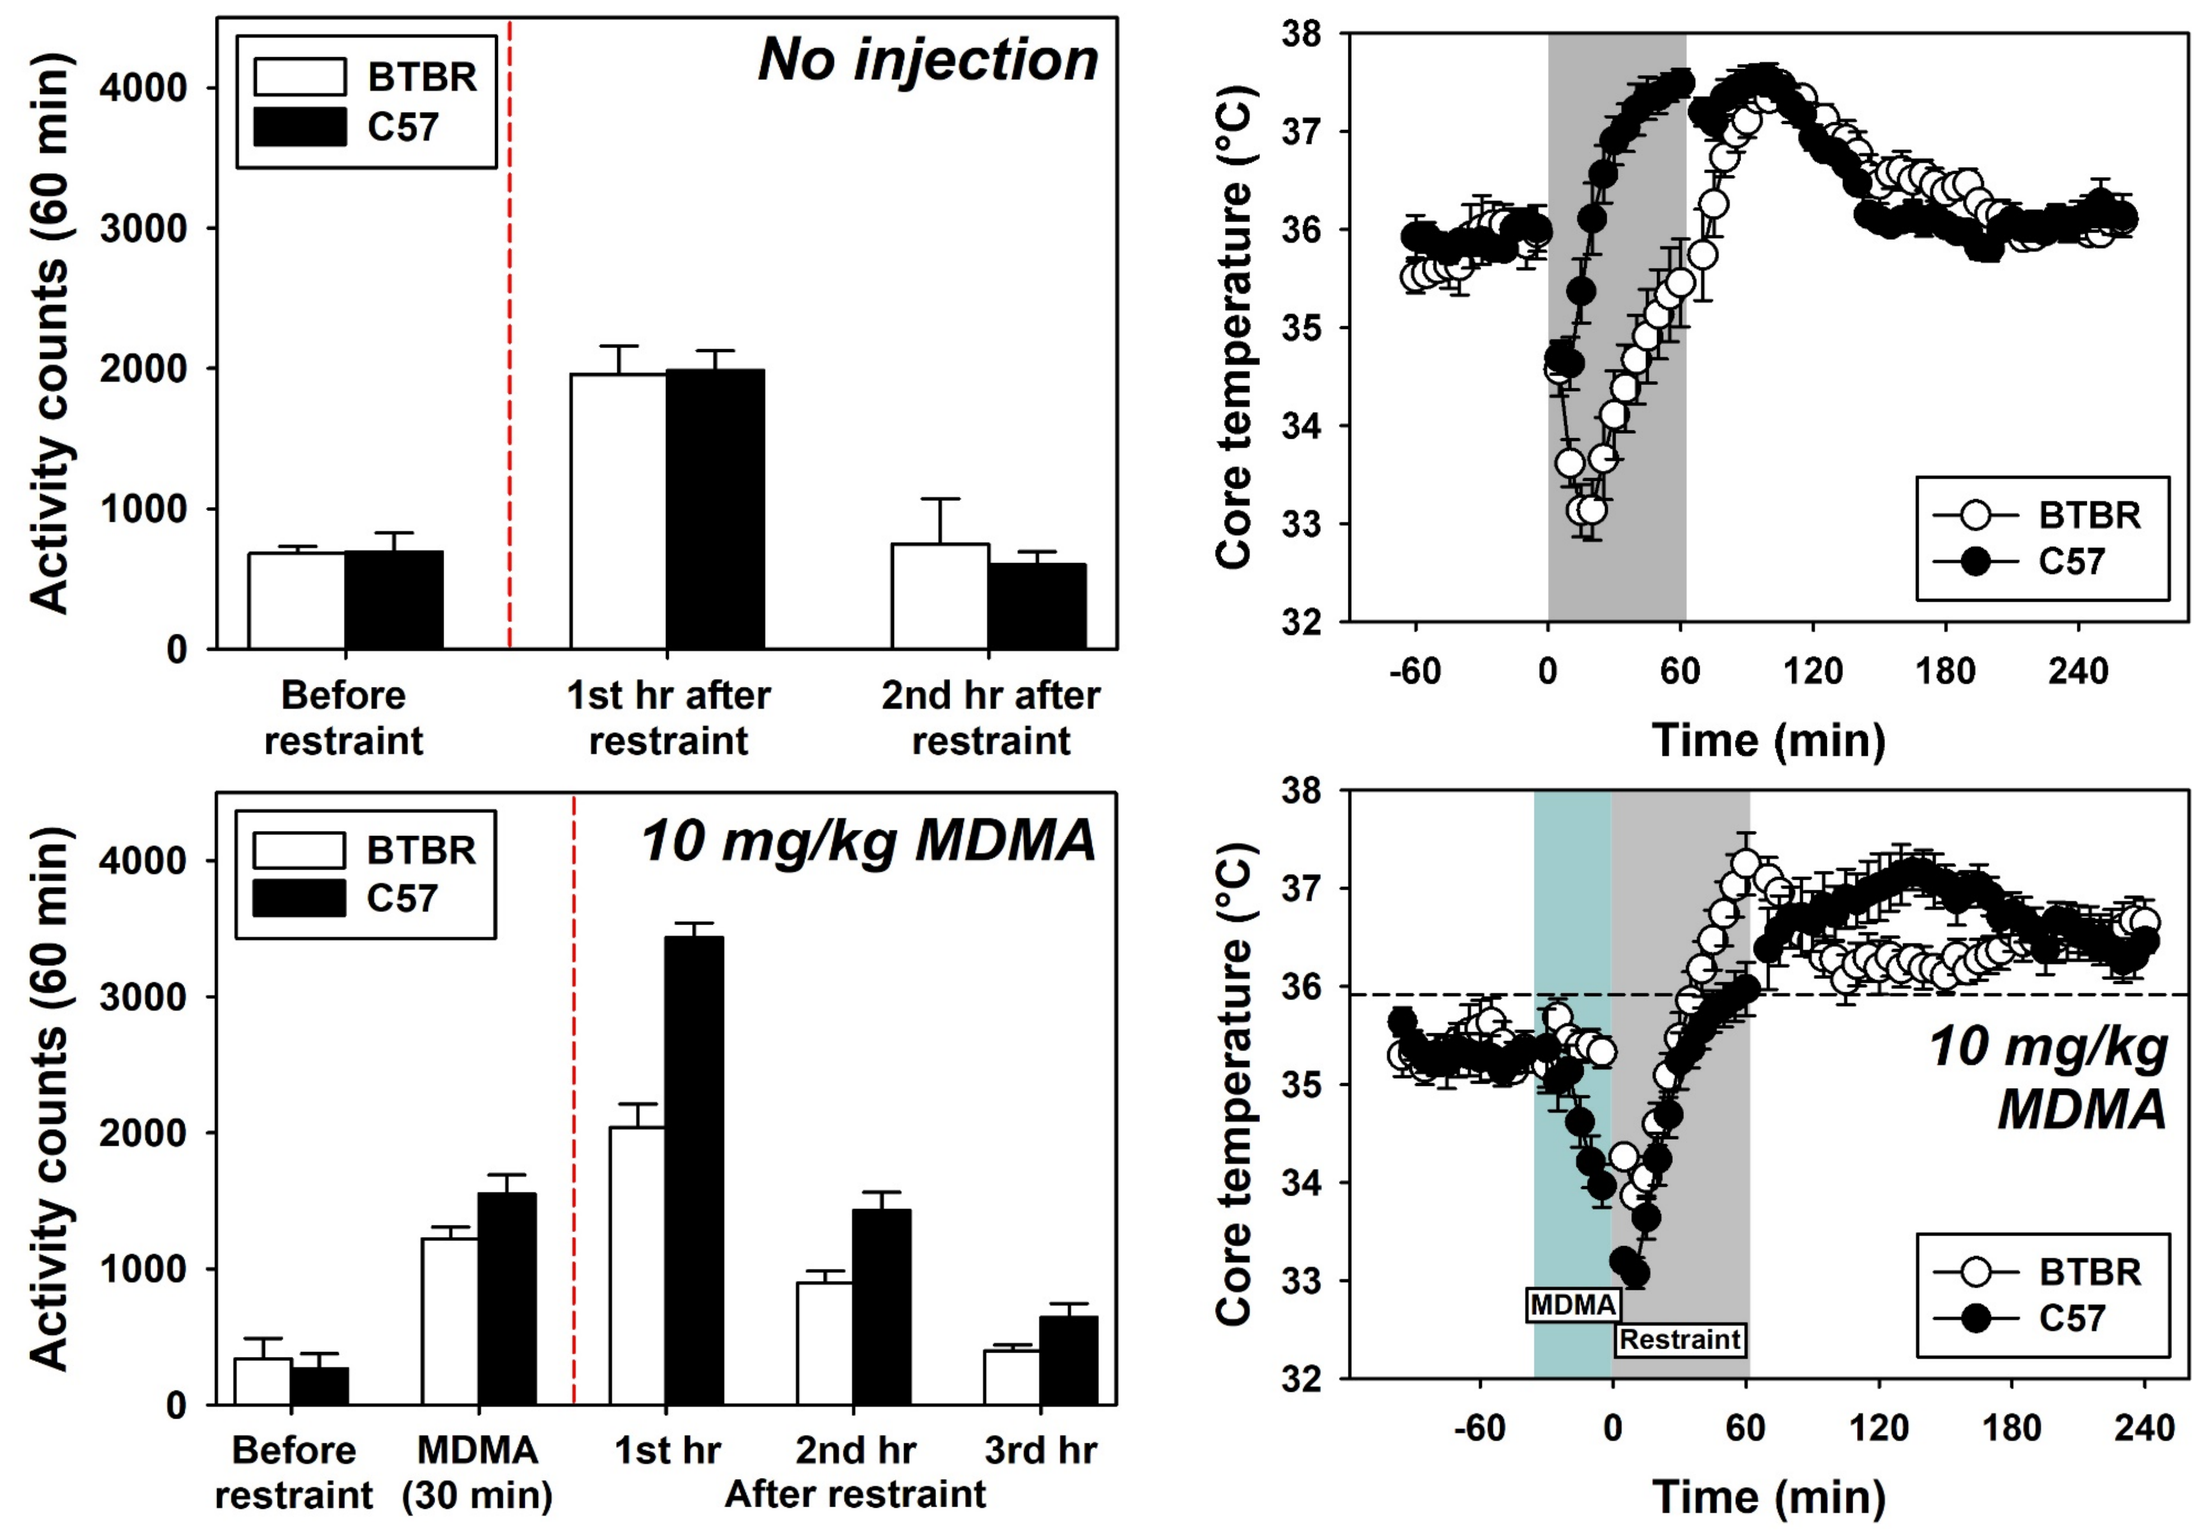

No injection
10 mg/kg MDMA
BTBR
C57
Activity counts (60 min)
Before restraint
1st hr after restraint
2nd hr after restraint
MDMA (30 min)
1st hr
2nd hr
3rd hr
After restraint
Core temperature (°C)
Time (min)
10 mg/kg MDMA
MDMA
Restraint

- Restraint rapidly elicited hypothermic effects in both strains, but temperatures rose more rapidly in C57s.
- Following restraint, both strains exhibited transient hyperthermic effects that resolved within ~2.5 hours.
- Both strains showed an equivalent increase in locomotor activity in the hour following restraint.
- 10 mg/kg MDMA reduced core temperature in C57 mice prior to restraint. During restraint, core temperatures rose at the same rate for both strains. Following restraint, C57s exhibited higher core temperatures, but both strains recovered within ~2.5 hours.
- MDMA increased motor activity prior to restraint to a similar degree in both strains. After restraint, motor activity was higher in C57s.

S-METH and MDMA

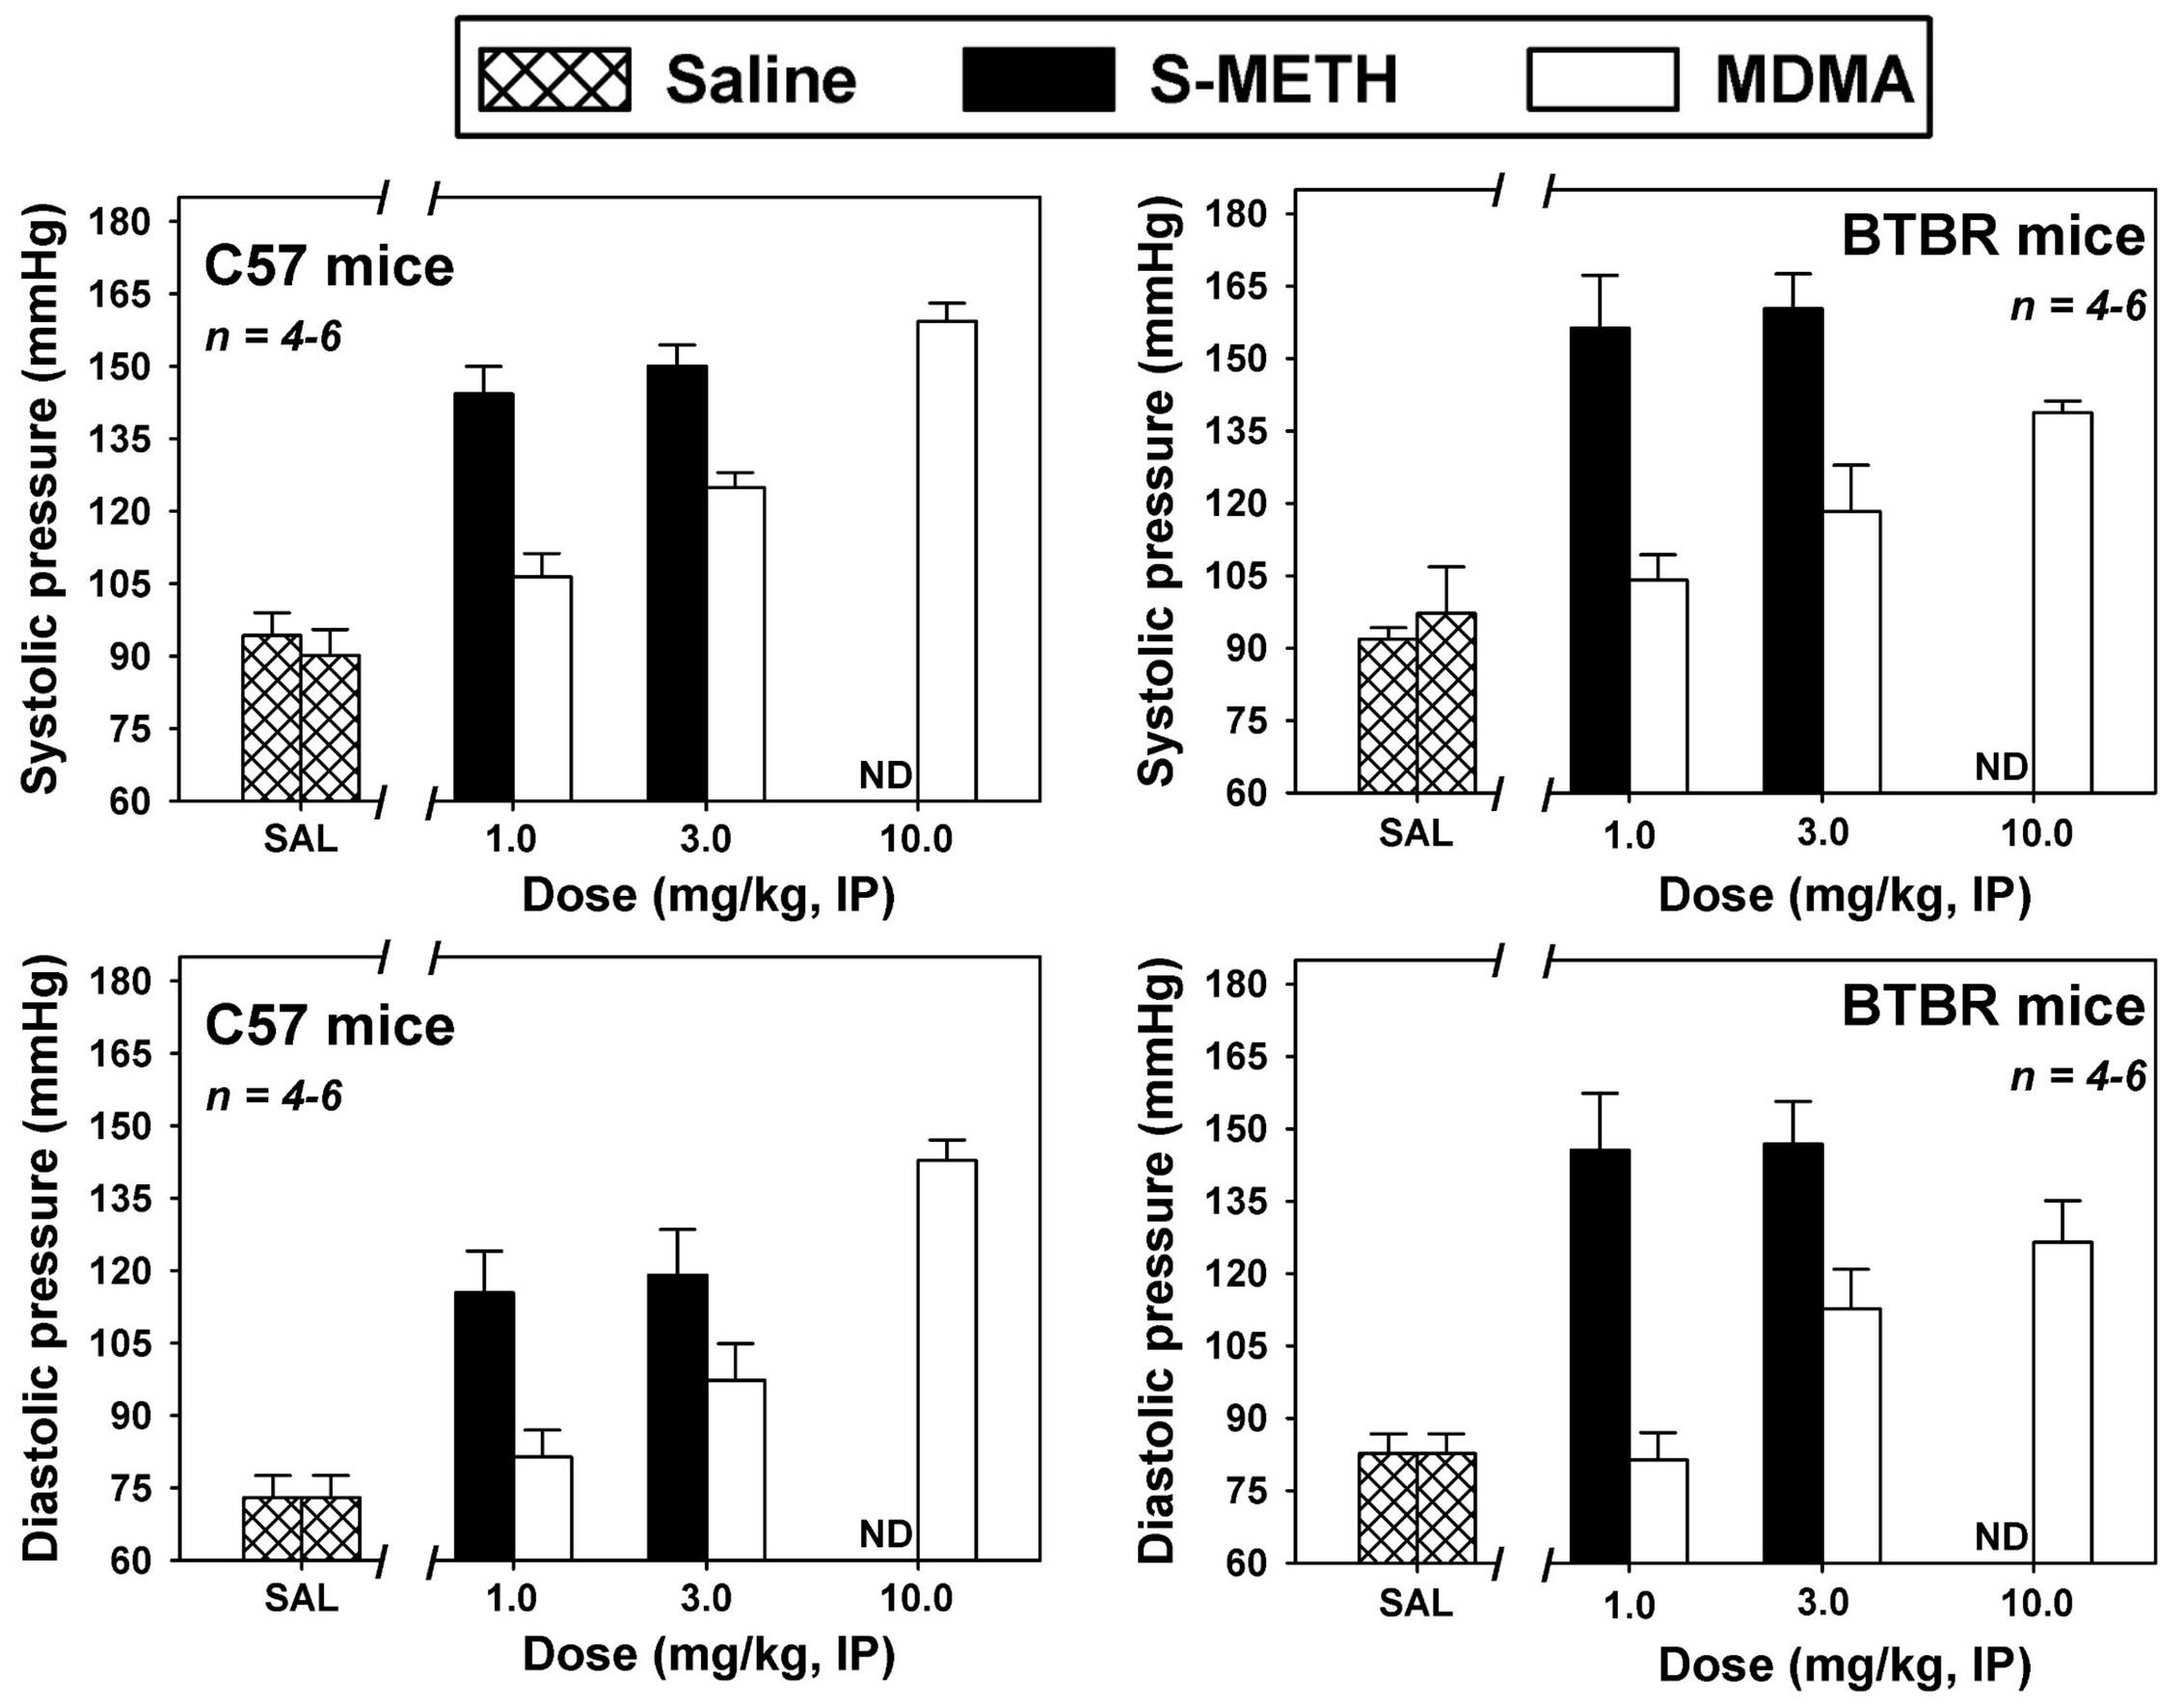

Saline
S-METH
MDMA
C57 mice
BTBR mice
n = 4-6
Systolic pressure (mmHg)
Diastolic pressure (mmHg)
SAL
1.0
3.0
10.0
ND
Dose (mg/kg, IP)

- S-METH and MDMA increased systolic and diastolic blood pressure in both C57 and BTBR mice.
- MDMA is less potent than S-METH.
- Strain differences are minor. S-METH is a bit more effective in BTBR mice, but MDMA is a bit less effective in BTBR mice.

5-EAPB and 6-EAPB

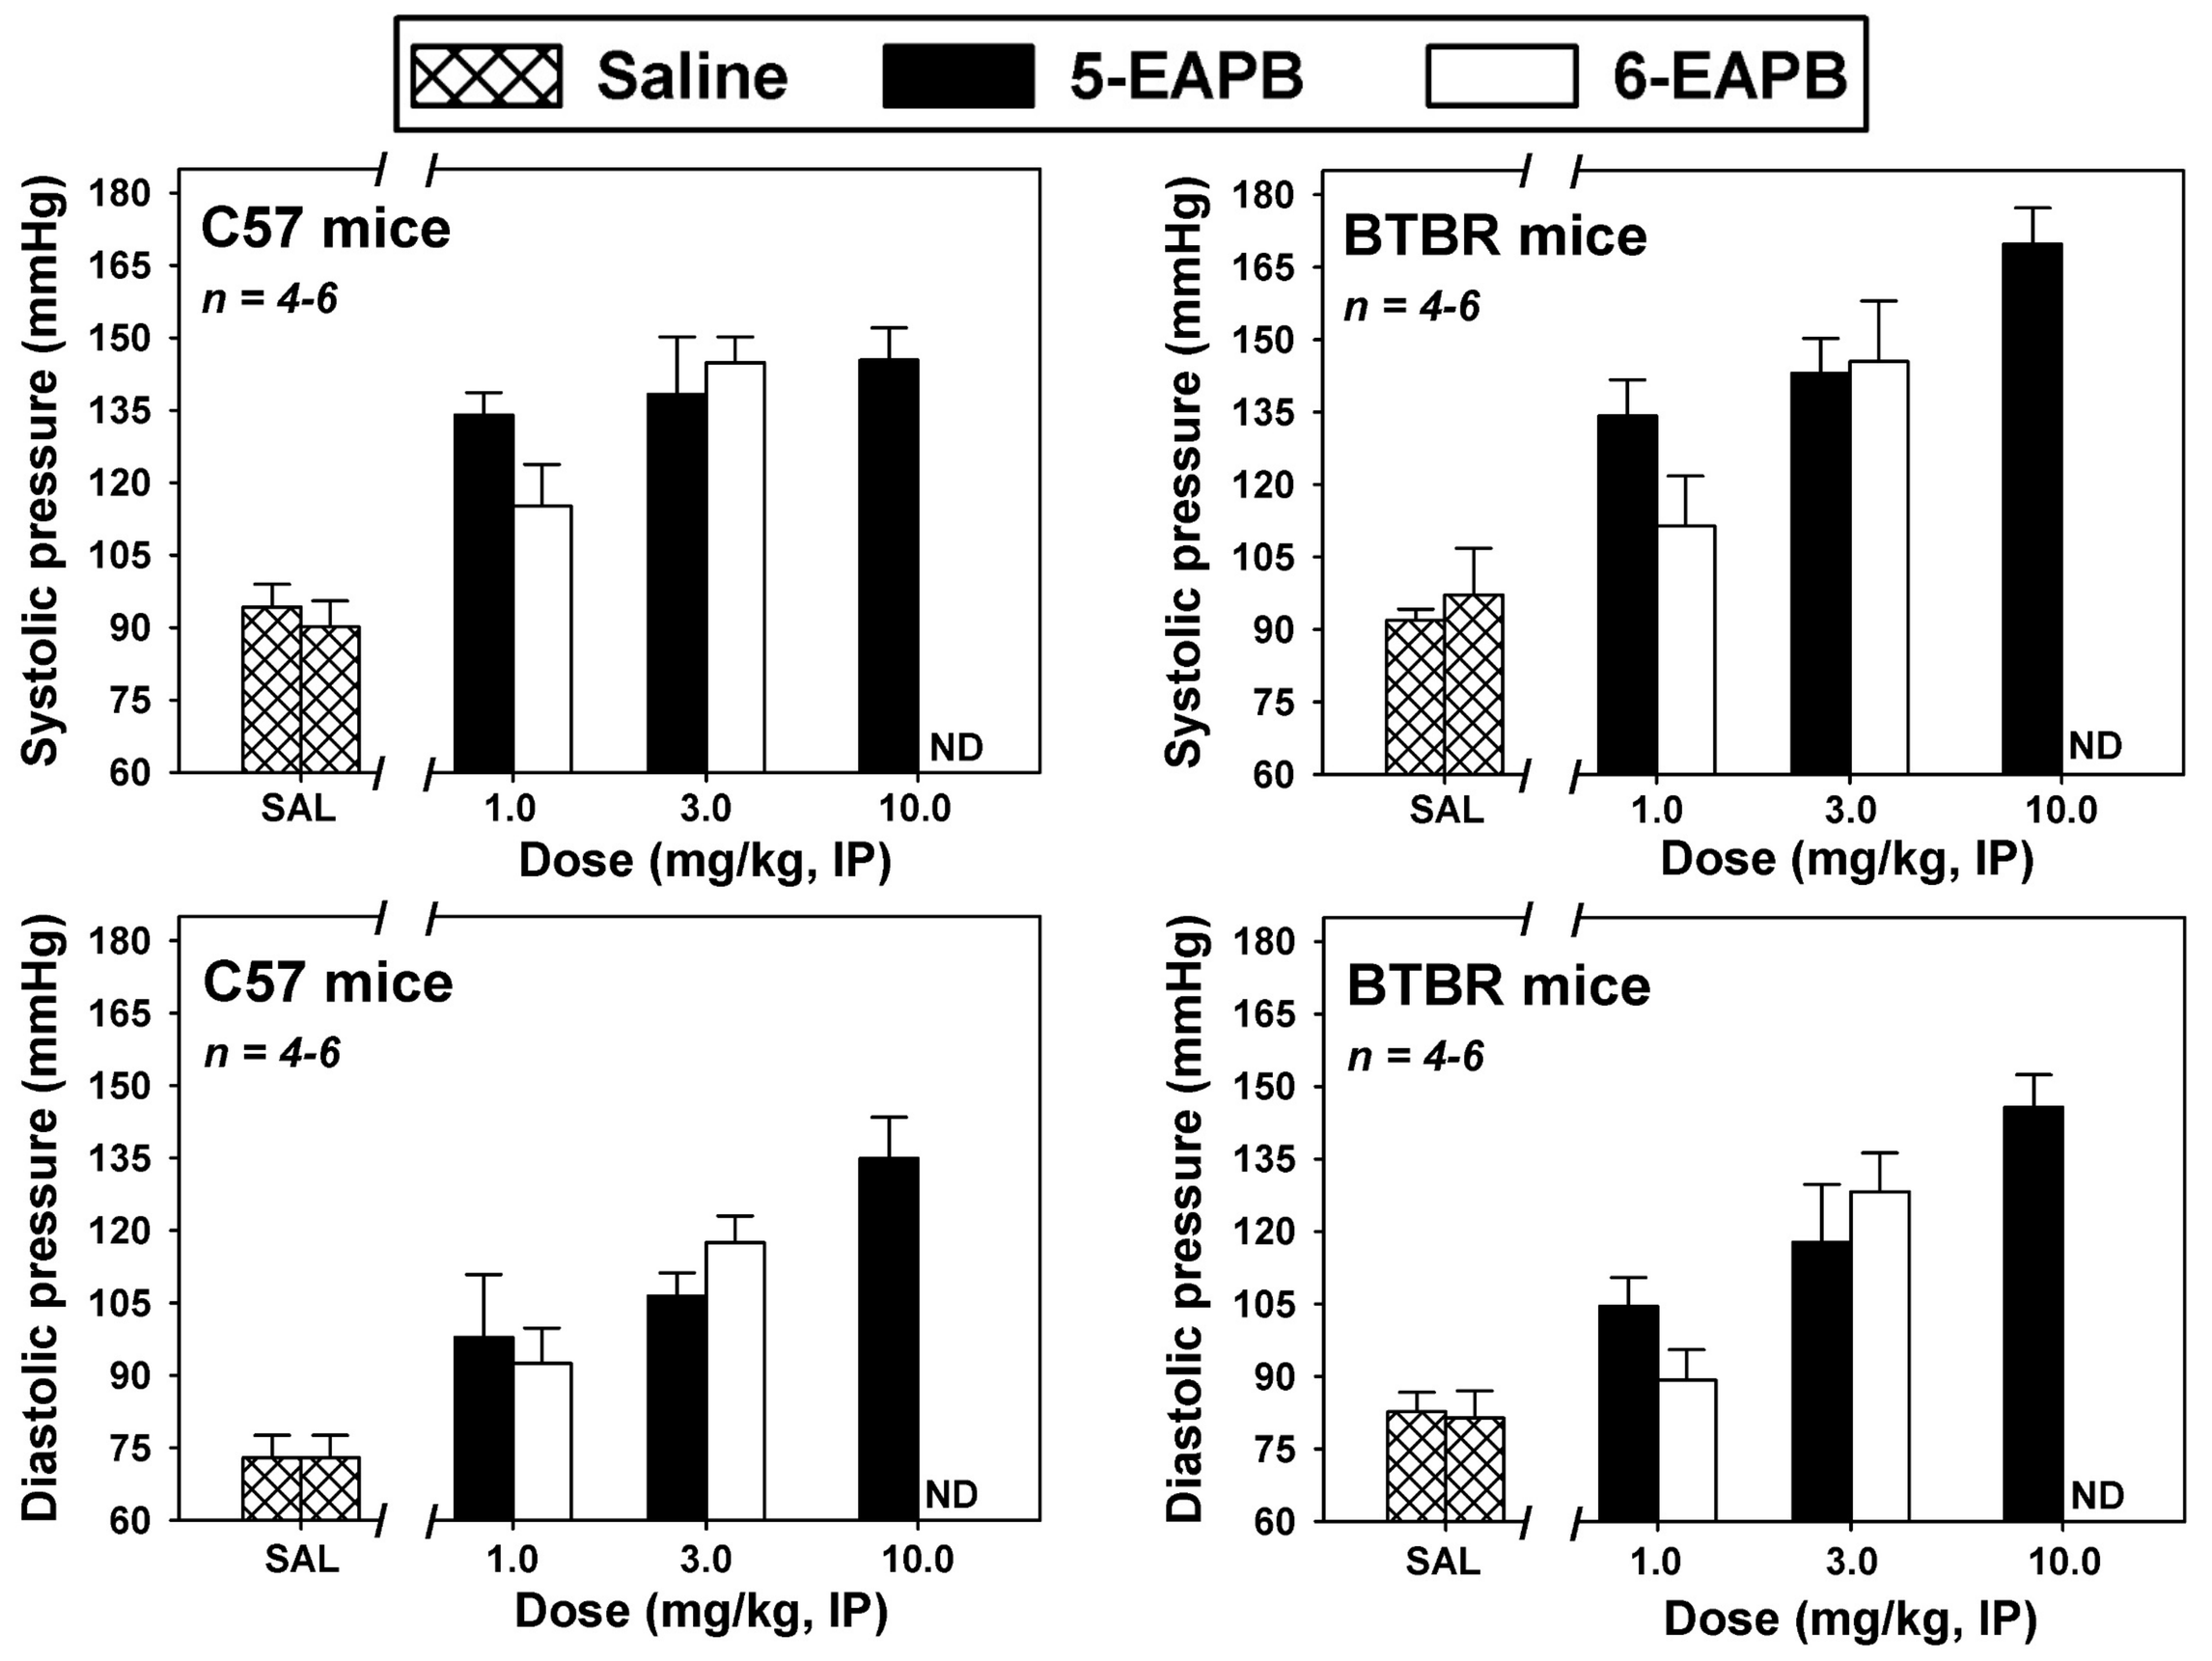

Saline
5-EAPB
6-EAPB
C57 mice
BTBR mice
n = 4-6
Systolic pressure (mmHg)
Diastolic pressure (mmHg)
SAL
1.0
3.0
10.0
ND
Dose (mg/kg, IP)

- 5-EAPB and 6-EAPB increased systolic and diastolic blood pressure in both C57 and BTBR mice.
- Similar potency and effectiveness across drugs and both EAPBs a bit more effective in BTBR mice.

CONCLUSIONS

- These studies reveal differences in restraint stress reactivity and thermoregulatory effects of MDMA between BTBR and C57 mice. Both of these factors could influence cardiovascular measurements using the tail cuff system.
- Effects of these four structurally-related drugs differed across strains in unpredictable ways. Although BTBR mice were perhaps more sensitive to cardiovascular effects of S-METH and the EAPBs, they were less sensitive to those of MDMA.
- All of the drugs tested produced adverse cardiovascular effects in both mouse strains. Drug development efforts to generate novel MDMA-like entactogens should focus on minimizing these effects on blood pressure.
- In vivo evaluations of new MDMA-like medications should determine therapeutic doses as well as doses which elicit adverse effects in order to better evaluate drug safety and commercial viability.

FUTURE DIRECTIONS

- Few studies have assessed drug effects in BTBR mice. The present results show that drug effects may not be predictable based on findings in the C57 progenitor strain. Effects of drugs from other pharmacological classes should be studied in the BTBR mouse.
- Novel non-racemic formulations of MDMA and related compounds may maintain therapeutic effects with reduced adverse effects. The effects of multiple R/S enantiomer ratios should be assessed.
- Strain differences between C57 and BTBR mice could be due to pharmacokinetics, pharmacodynamics, or both. Comparative drug metabolism and 5-HT receptor / transporter studies should be performed.

ACKNOWLEDGEMENTS

- These studies funded in part by PharmAla Biotech and by a graduate fellowship to KHH through T32 DA022981 *Translational training in addiction.*